Acoramidis Produces Near-Complete TTR Stabilization in Blood Samples from Patients with Variant Transthyretin Amyloidosis that is Greater than that Achieved with Tafamidis

Alan X Ji[1], Paul Wong[1], Daniel P Judge[2], Isabella A Graef[3], Jonathan Fox[1], and Uma Sinha[1].

[1]Eidos Therapeutics, Inc. a BridgeBio Company. [2]Medical University of South Carolina. [3]Stanford University.

INTRODUCTION

- Transthyretin (TTR) amyloidosis (ATTR) is a progressive, fatal disease caused by destabilizing TTR variants (TTRv) and age-related factors.
- Deposition of TTR amyloid causes severe organ damage and dysfunction.
- Familial transthyretin amyloid cardiomyopathy (ATTRv-CM) and amyloid polyneuropathy (ATTR-PN), are driven by pathogenic point mutations in the ATTR gene; over 100 such variants have been described. More destabilizing variants are associated with more severe clinical phenotypes.
- Dissociation of the TTR tetramer into its constituent monomers is the rate-limiting step in amyloidogenesis. TTR stabilizers have demonstrated clinical benefits for neuropathic and cardiovascular outcomes correlated with the extent of TTR stabilization.
- Acoramidis (AG10) is a novel TTR stabilizer in development for the treatment of ATTR-CM. Acoramidis binds the tetrameric form of TTR and prevents its dissociation into dimers and monomers.
- Objectives:
 - Determine if acoramidis at its target steady-state trough concentration can demonstrate pan-variant potency.
 - Directly compare in vitro tetramer stabilization by acoramidis against tafamidis, a TTR stabilizer in clinical use.
 - Determine if in vitro ATTRv stabilization assay findings correlate with in vivo changes in serum TTR observed in the recently completed Phase 3 ATTRibute-CM study of acoramidis.

HYPOTHESIS

- Acoramidis is a highly selective and potent stabilizer of TTR.
- Acoramidis achieves near-complete in vitro stabilization, exceeding levels achieved with clinically relevant concentrations of tafamidis, when added to blood samples from ATTRv patients across a spectrum of destabilizing TTR variants.

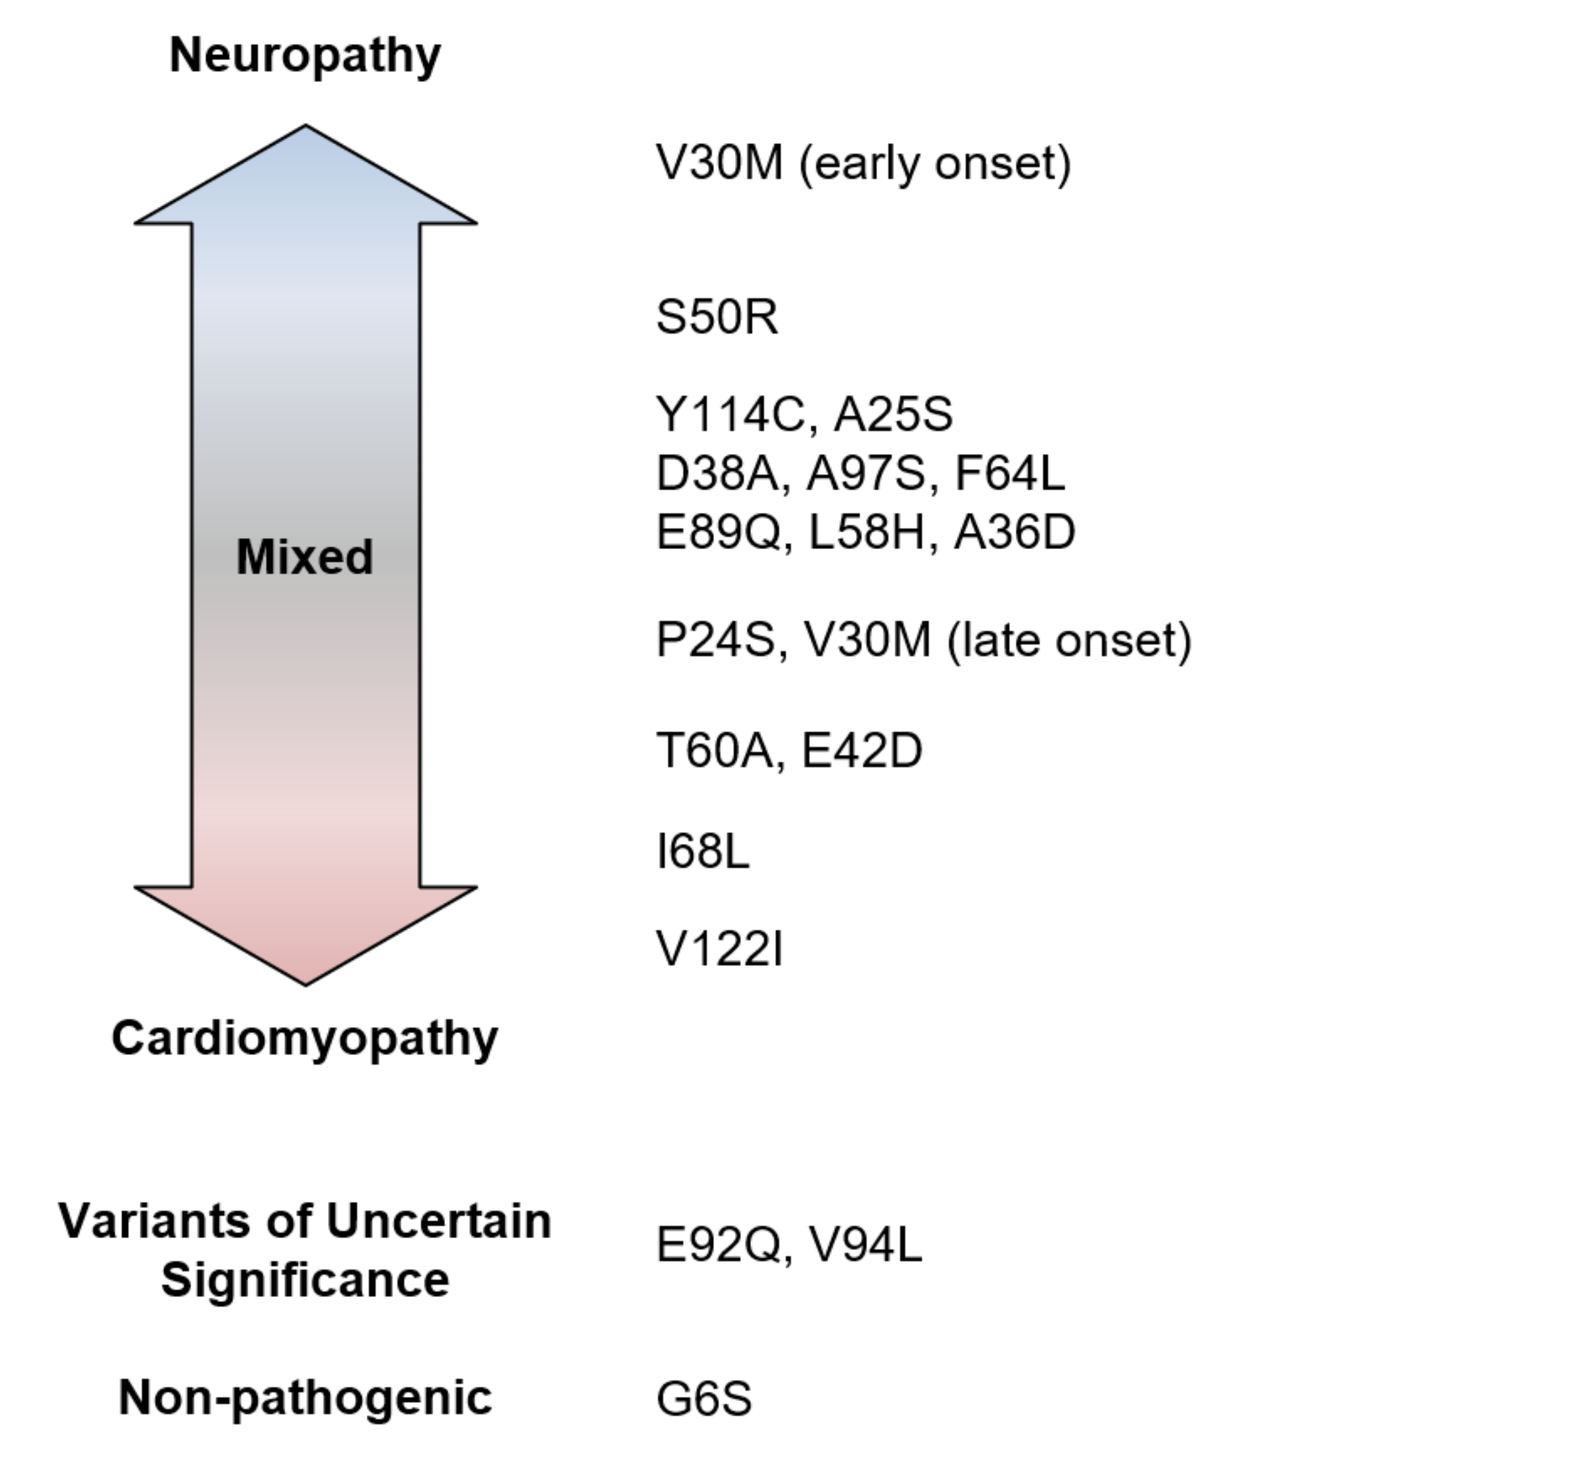
Neuropathy
Mixed
Cardiomyopathy
V30M (early onset)
S50R
Y114C, A25S
D38A, A97S, F64L
E89Q, L58H, A36D
P24S, V30M (late onset)
T60A, E42D
I68L
V122I
Variants of Uncertain Significance
E92Q, V94L
Non-pathogenic
G6S

Figure 1. *Spectrum of TTR variants and related symptoms[1,2].*

METHODS

- Two established pharmacodynamic assays[3] measure the stabilization of TTR tetramers in vitro: Western Blot (WB), and Fluorescent Probe Exclusion (FPE)
 - The WB assay quantifies intact TTR tetramer after acid mediated accelerated dissociation.
 - The FPE assay uses a highly specific and selective fluorescent probe to measure TTR ligand binding site occupancy.
- Blood samples were collected from subjects with TTR variants screened for the Phase 3 ATTRibute-CM clinical trial and other acoramidis clinical studies.
- Over 60 individual patient samples representing 18 unique TTRv across a spectrum of intrinsic instability and clinical phenotypes were assayed.
- Acoramidis was added to patient samples at 10 μM concentration. The target clinical steady-sate trough concentration is 8-10 μM.
- Tafamidis was added to patient samples at its clinical peak (26 μM) and trough (16 μM) concentrations reported for its maximal approved dose[4].
- WB Assay:
 - Samples are acidified for 72 hours. Samples are then crosslinked and resolved by SDS-PAGE. The ratio of TTR tetramer in the acidified sample is compared to the amount of tetramer in the original sample.
 - $WB\ \%Stabilization = 100 \times \frac{72\ hr\ TTR\ tetramer}{0\ hr\ TTR\ tetramer}$
- FPE Assay:
 - A fluorescent probe is added to each sample which only fluoresces upon covalently binding to the ligand binding site of TTR. The presence of acoramidis or tafamidis in the binding site suppresses the development of fluorescent signal. The target engagement for each stabilizer is calculated at the 1-hour timepoint. Note: DMSO = dimethyl sulfoxide.
 - $FPE\ \%Stabilization_X = 100 \times \frac{(FPE_{DMSO}\ @60\ min)-(FPE_X\ @60\ min)}{(FPE_{DMSO}\ @60\ min)}$

ACORAMIDIS MORE EFFECTIVELY STABILIZES VARIANT TTR THAN TAFAMIDIS

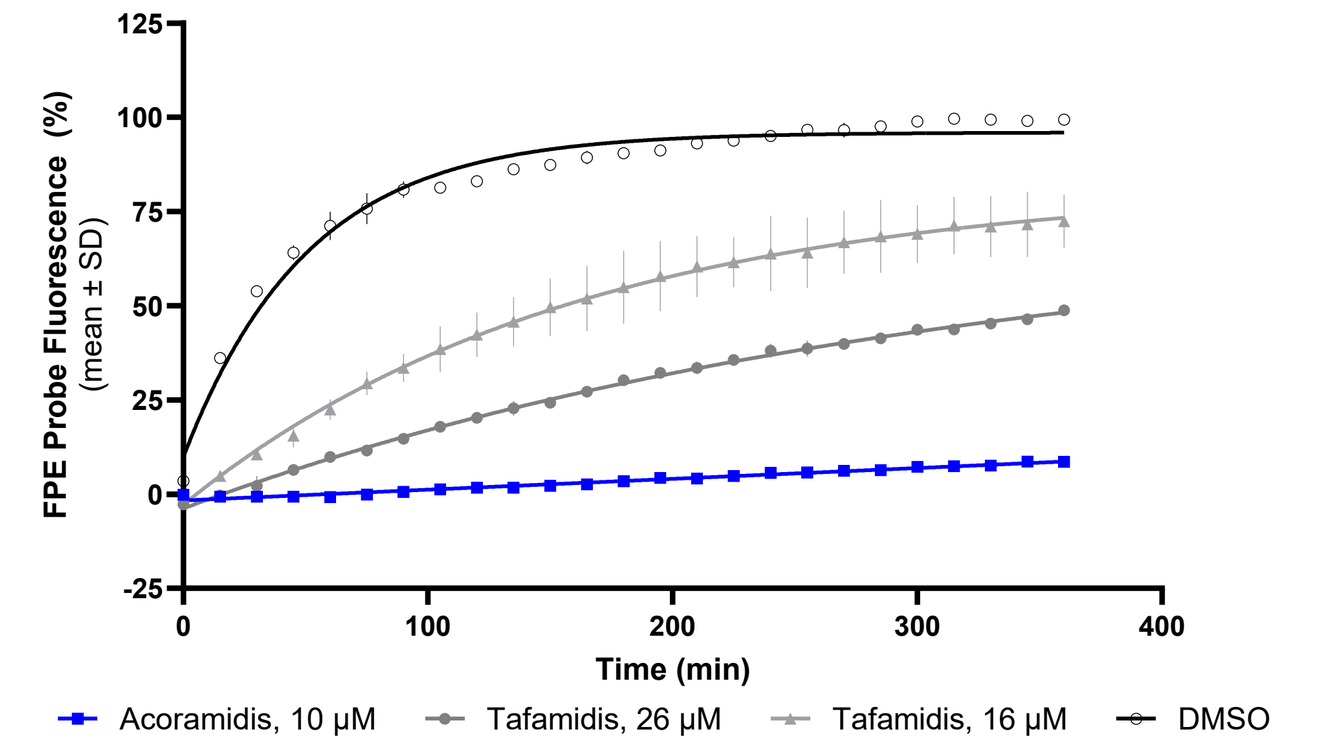
FPE Probe Fluorescence (%) (mean ± SD)
Time (min)
Acoramidis, 10 μM
Tafamidis, 26 μM
Tafamidis, 16 μM
DMSO

Figure 2. *FPE time course of an individual V122I patient sample.*

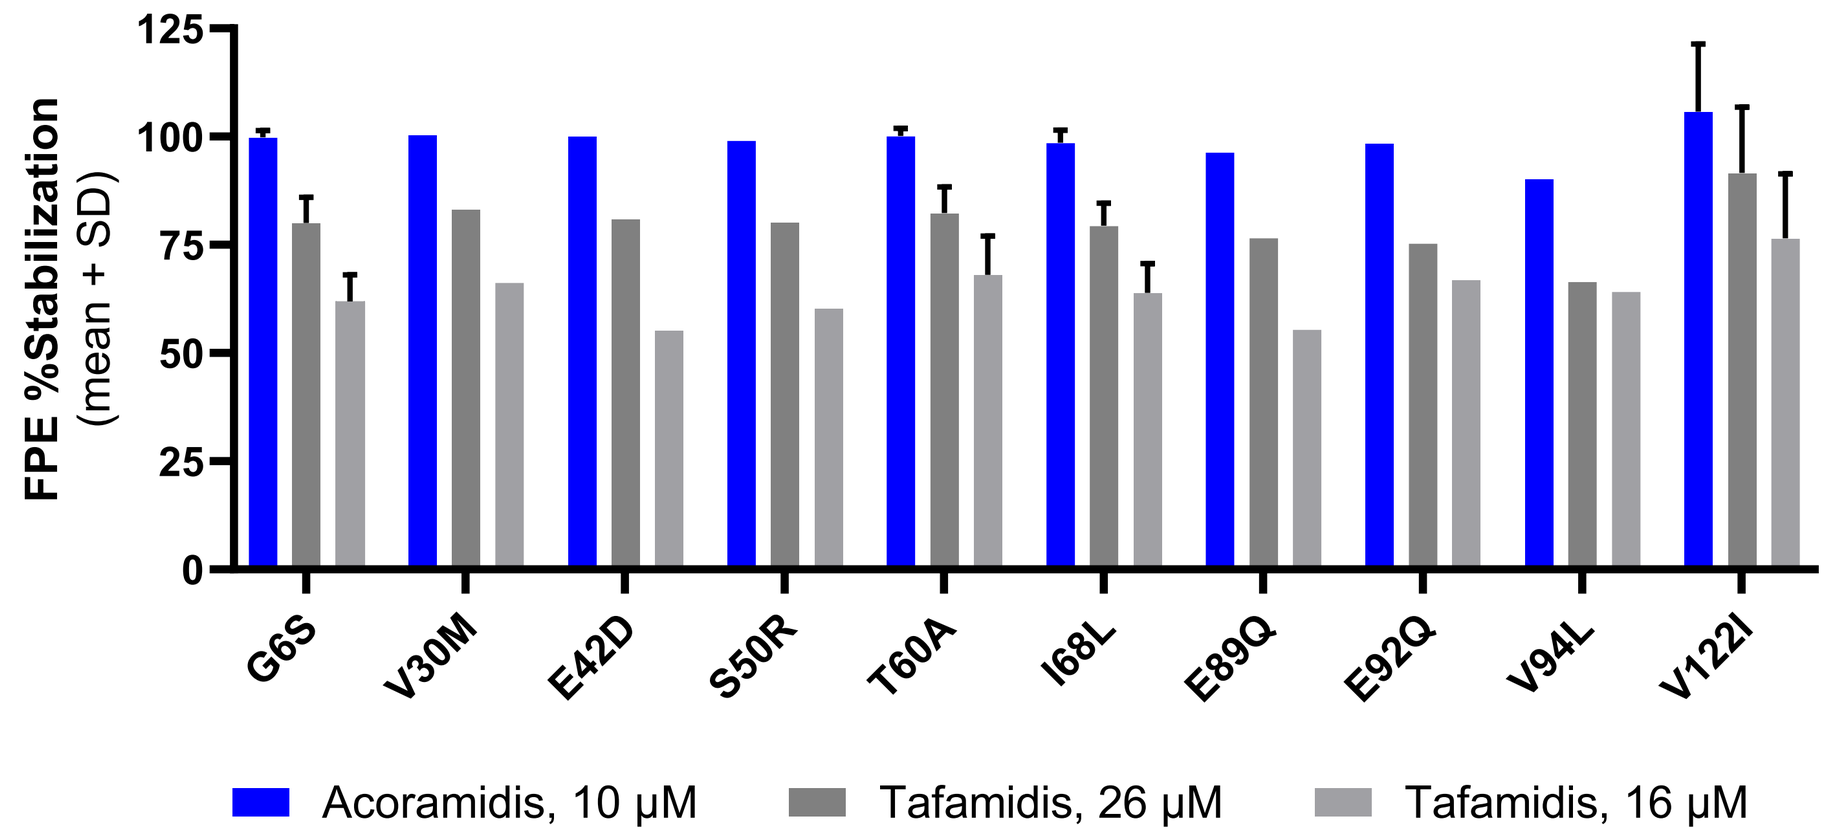
FPE %Stabilization (mean + SD)
G6S
V30M
E42D
S50R
T60A
I68L
E89Q
E92Q
V94L
V122I
Acoramidis, 10 μM
Tafamidis, 26 μM
Tafamidis, 16 μM

Figure 3. *TTR FPE %Stabilization by variant. SD is only shown for measurements with 2 or more samples.*

ACORAMIDIS DEMONSTRATED PAN-VARIANT ACTIVITY TO A GREATER EXTENT THAN TAFAMIDIS

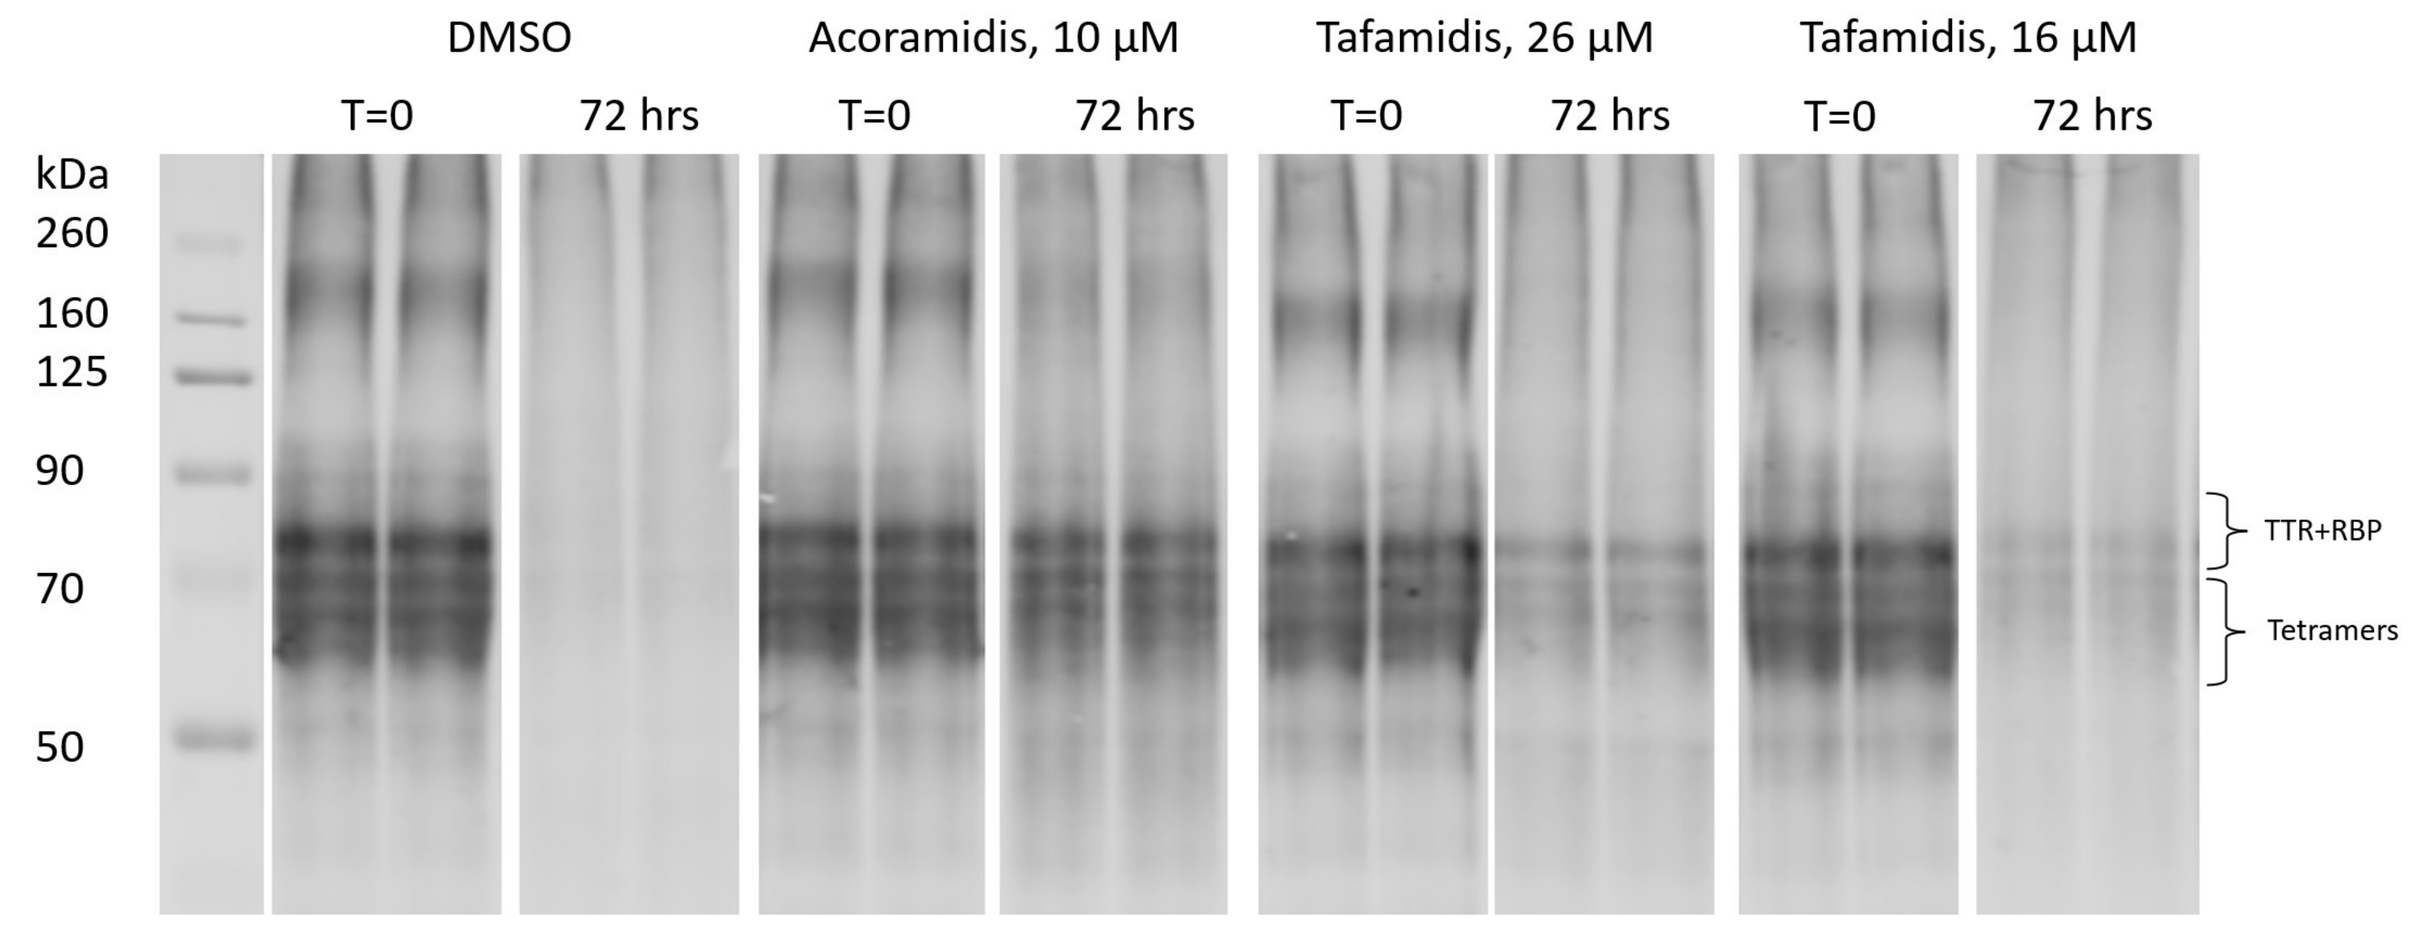
DMSO
Acoramidis, 10 μM
Tafamidis, 26 μM
Tafamidis, 16 μM
T=0
72 hrs
kDa
260
160
125
90
70
50
TTR+RBP
Tetramers

Figure 4. *WB of an individual V122I patient sample. All conditions were run in duplicate lanes. Brackets denoting the bands corresponding to tetrameric TTR with or without Retinol Binding Protein (RBP) are indicated.*

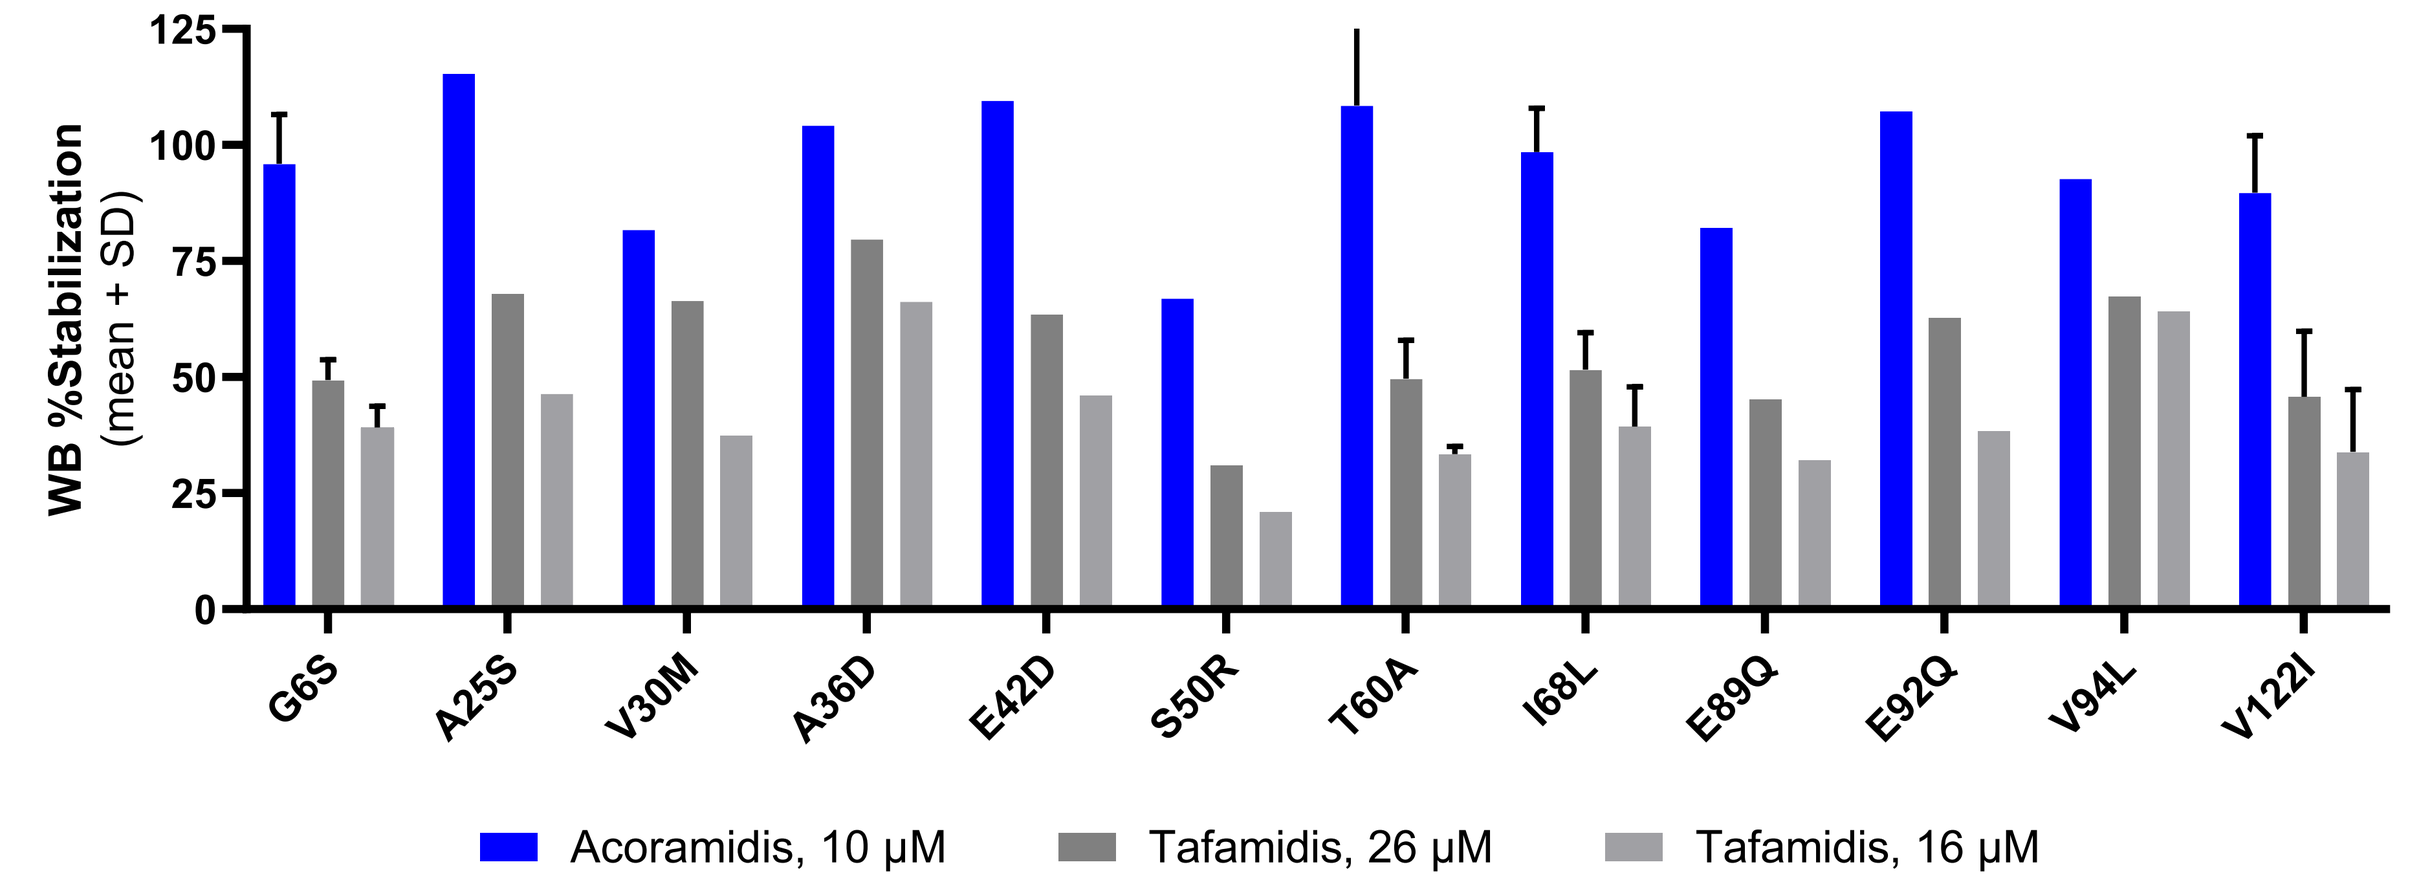
WB %Stabilization (mean + SD)
G6S
A25S
V30M
A36D
E42D
S50R
T60A
I68L
E89Q
E92Q
V94L
V122I
Acoramidis, 10 μM
Tafamidis, 26 μM
Tafamidis, 16 μM

Figure 5. *TTR WB %Stabilization by variant. SD is only shown for measurements with 2 or more samples.*

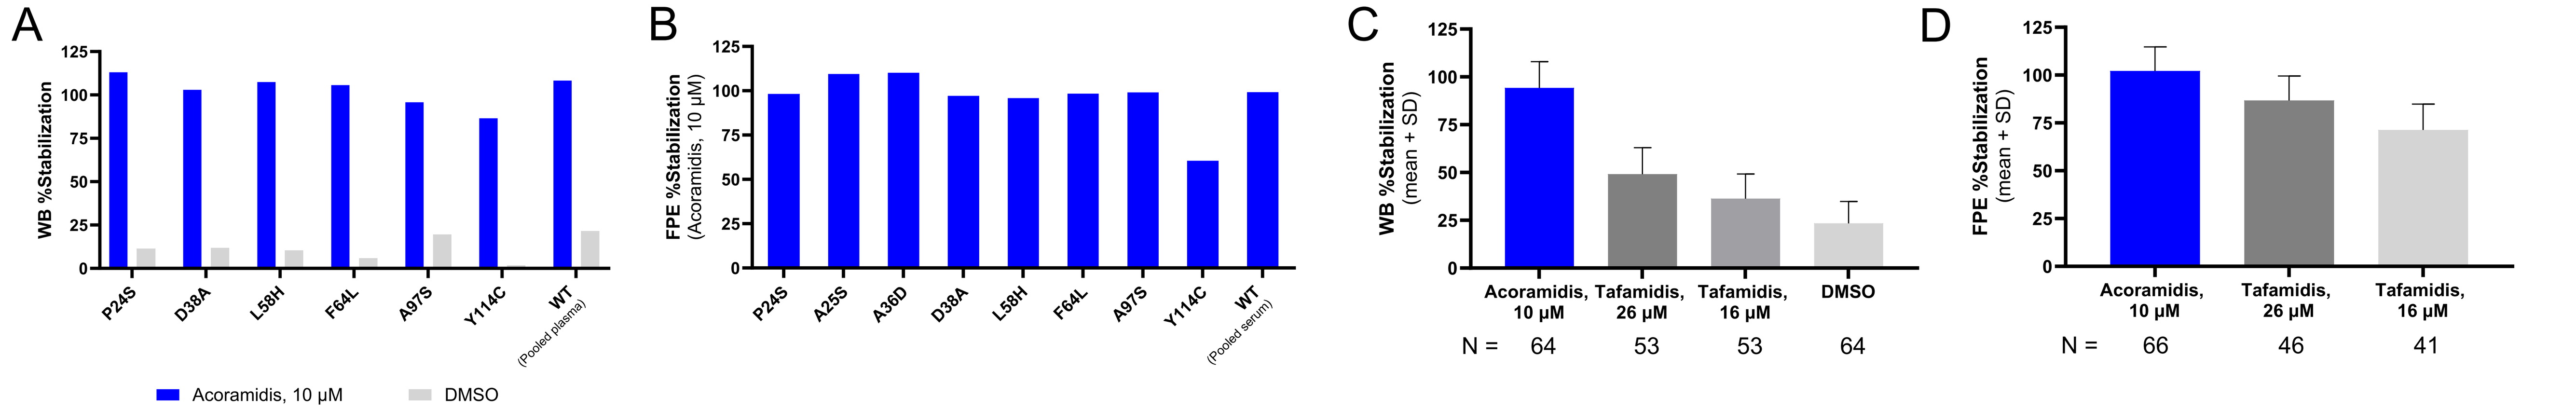
A
B
C
D
WB %Stabilization
P24S
D38A
L58H
F64L
A97S
Y114C
WT (Pooled plasma)
Acoramidis, 10 μM
DMSO
FPE %Stabilization (Acoramidis, 10 μM)
P24S
A25S
A36D
D38A
L58H
F64L
A97S
Y114C
WT (Pooled serum)
WB %Stabilization (mean + SD)
Acoramidis, 10 μM
Tafamidis, 26 μM
Tafamidis, 16 μM
DMSO
N = 64 53 53 64
FPE %Stabilization (mean + SD)
Acoramidis, 10 μM
Tafamidis, 26 μM
Tafamidis, 16 μM
N = 66 46 41

Figure 6. *Stabilization of unique variant patient samples measured by **A)** WB and **B)** FPE, with wildtype (WT) results as reference. Overall stabilization across all tested variant samples measured by **C)** WB and **D)** FPE.*

Table 1. Summary of head-to-head WB %Stabilization Results

Variant	N	WB Mean %Stabilization (SD): Acoramidis, 10 μM	Tafamidis, 26 μM	Tafamidis, 16 μM	DMSO
G6S	2	95.93 (10.67)	49.35 (4.37)	39.16 (4.59)	28.27 (0.13)
A25S	1	115.43	68.06	46.47	20.88
V30M	1	81.73	66.51	37.47	31.95
A36D	1	104.20	79.70	66.29	50.54
E42D	1	109.63	63.57	46.08	24.14
S50R	1	67.00	31.00	21.00	12.00
T60A	3	108.51 (25.51)	49.64 (8.33)	33.44 (1.60)	23.41 (6.58)
I68L	7	98.53 (9.39)	51.6 (8.00)	39.34 (8.59)	21.38 (7.61)
E89Q	1	82.23	45.26	32.16	26.96
E92Q	1	107.21	62.76	38.49	25.89
V94L	1	92.79	67.37	64.19	25.96
V122I	33	89.70 (12.33)	45.73 (14.13)	33.93 (13.39)	25.50 (12.97)
Variant mean	**53**	**92.97 (13.97)**	**49.10 (13.89)**	**36.34 (12.88)**	**25.21 (11.46)**

Note: SD is only shown for measurements with 2 or more samples. Overall mean WB %Stabilization by 10 μM acoramidis is significantly higher than 26 μM tafamidis ($p<0.0001$).

Table 2. Summary of head-to-head FPE %Stabilization Results

Variant	N	FPE Mean %Stabilization (SD): Acoramidis, 10 μM	Tafamidis, 26 μM	Tafamidis, 16 μM
G6S	2	99.81 (1.55)	80.02 (5.99)	61.96 (6.12)
V30M	1	100.34	83.15	66.32
E42D	1	100.05	80.99	55.28
S50R	1	99.11	80.27	60.34
T60A	4	100.12 (1.77)	82.32 (6.05)	68.10 (8.92)
I68L	5	98.55 (2.98)	79.4 (5.28)	63.95 (6.73)
E89Q	1	96.44	76.66	55.48
E92Q	1	98.46	75.41	66.97
V94L	1	90.13	66.46	64.22
V122I	24	105.76 (15.65)	91.62 (15.18)	76.54 (14.83)
Variant mean	**41**	**102.82 (12.52)**	**86.54 (13.51)**	**71.26 (13.58)**
Pooled serum (WT)	6*	99.37 (1.77)	64.69 (5.52)	48.61 (6.43)

Note: SD is only shown for measurements with 2 or more samples. Overall mean FPE %Stabilization by 10 μM acoramidis is significantly higher than 26 μM tafamidis ($p<0.0001$).
*Healthy pooled serum was tested using 6 replicates per assay condition.

EX VIVO TTR STABILIZATION CORRELATES WITH IN VIVO MEASUREMENT OF SERUM TTR

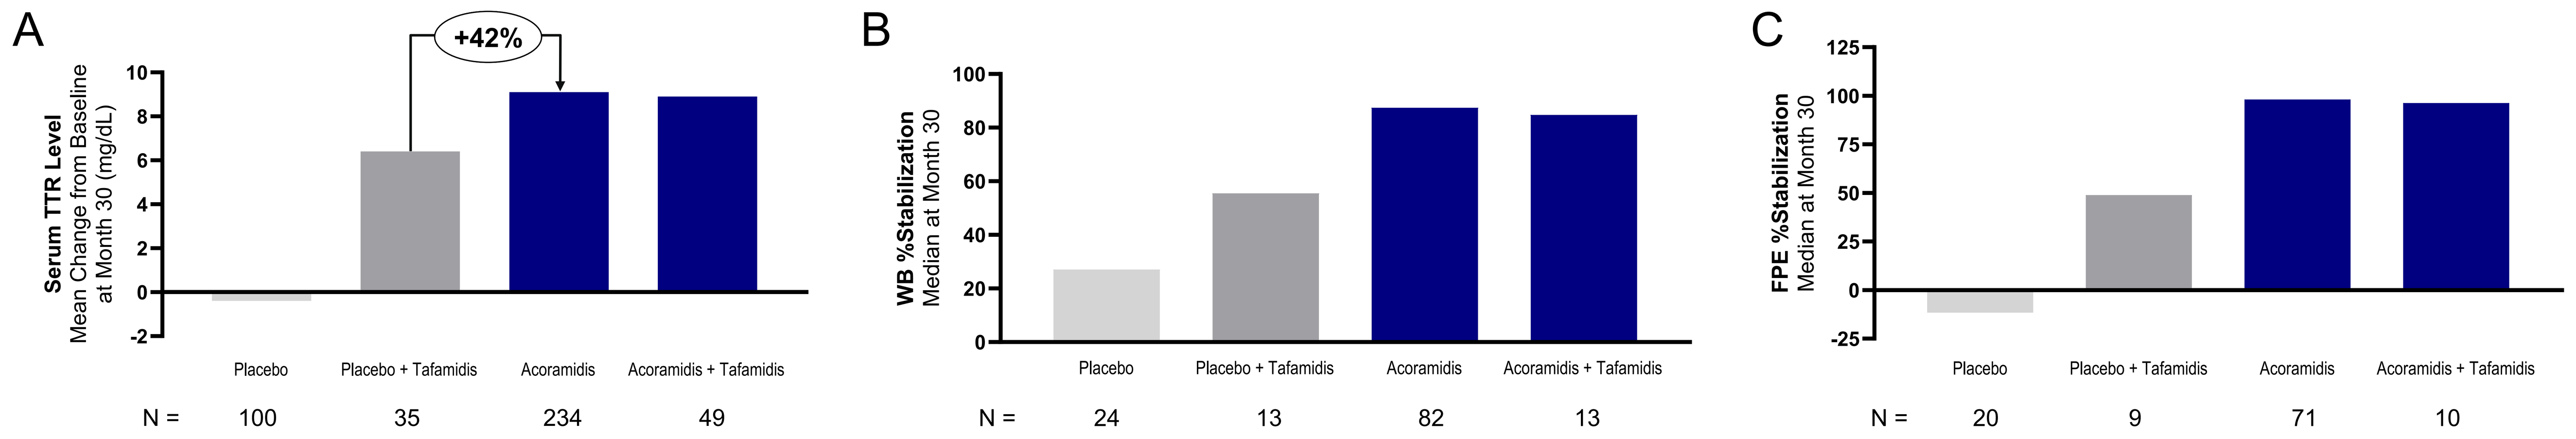
A
Serum TTR Level
Mean Change from Baseline at Month 30 (mg/dL)
+42%
Placebo
Placebo + Tafamidis
Acoramidis
Acoramidis + Tafamidis
N = 100 35 234 49
B
WB %Stabilization
Median at Month 30
N = 24 13 82 13
C
FPE %Stabilization
Median at Month 30
N = 20 9 71 10

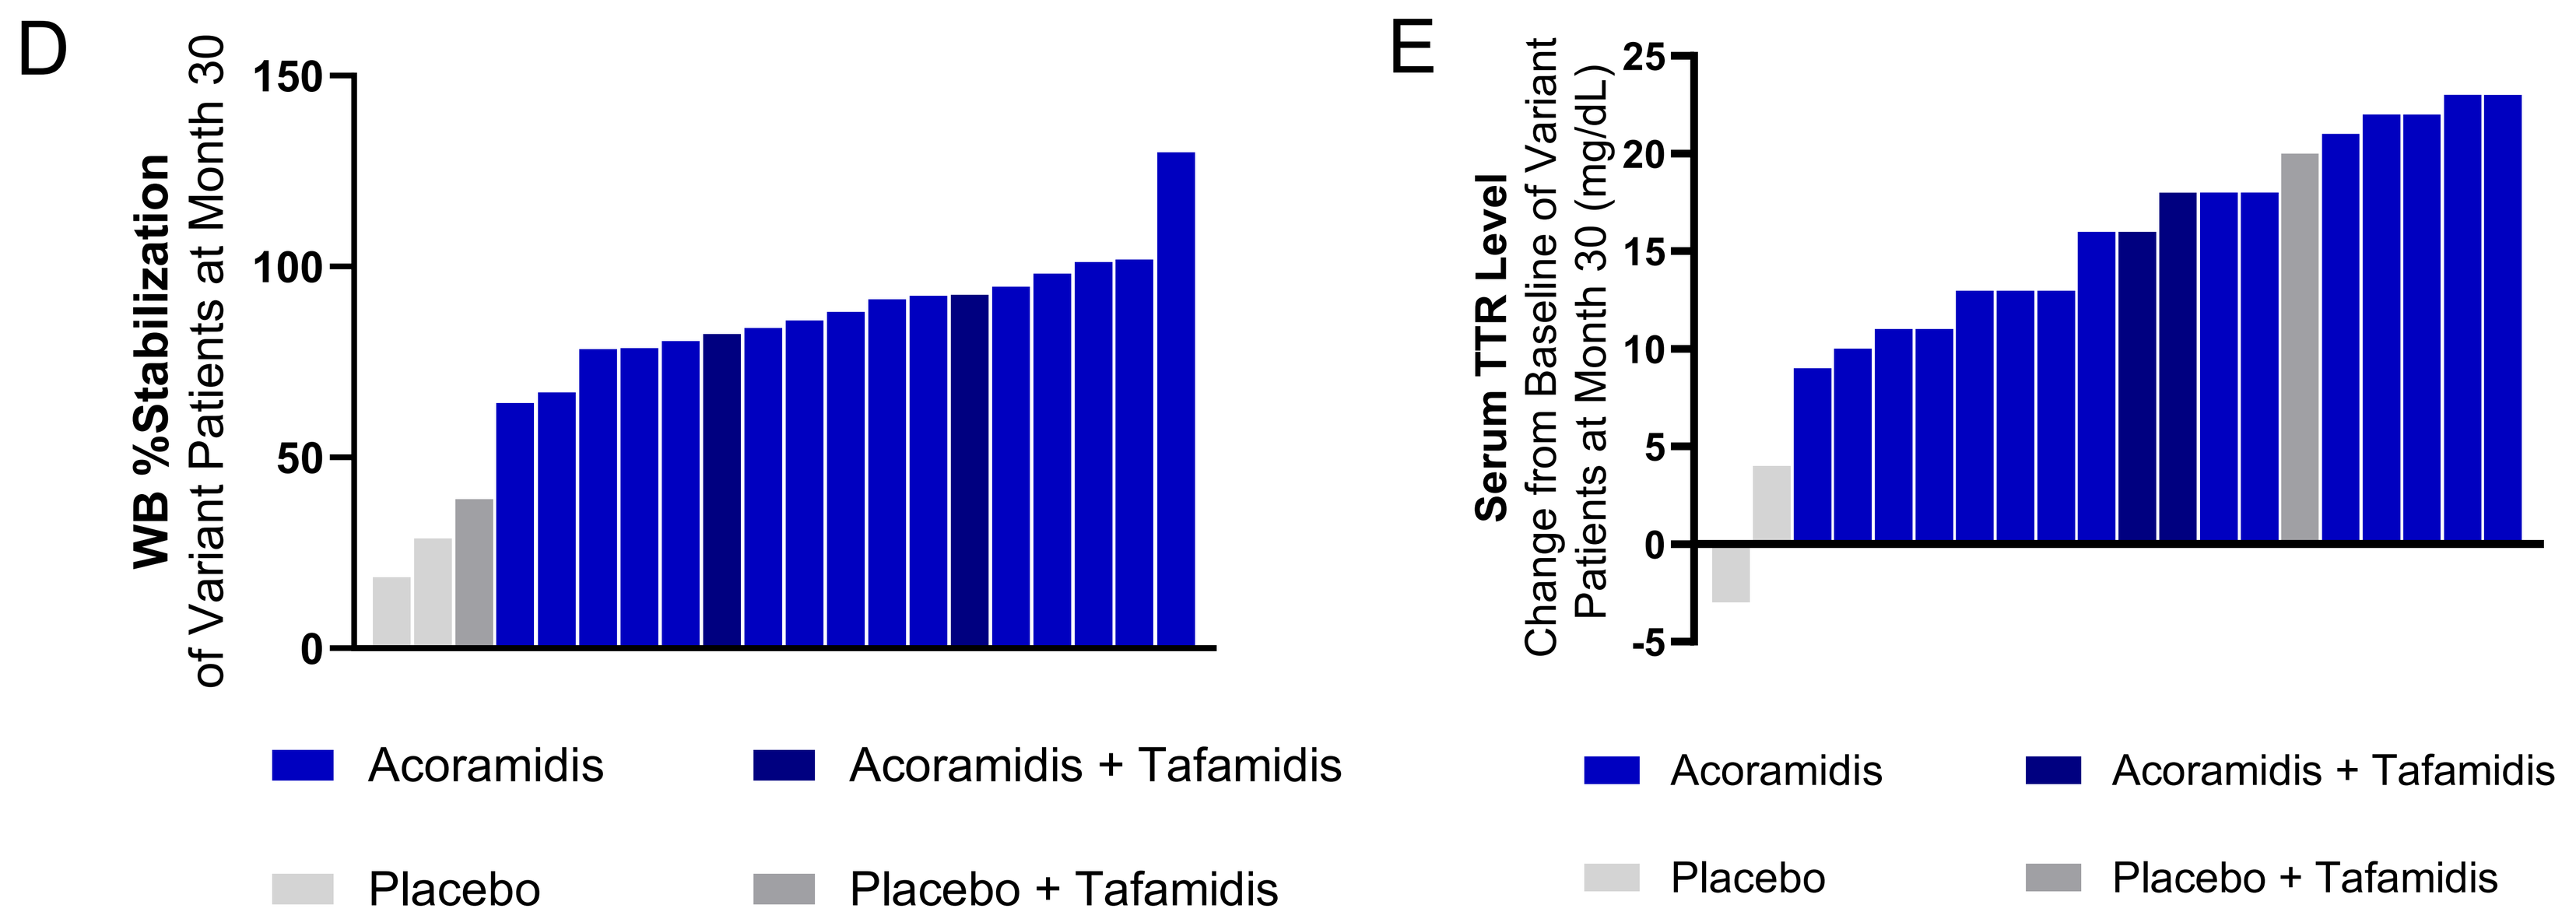
D
WB %Stabilization of Variant Patients at Month 30
E
Serum TTR Level
Change from Baseline of Variant Patients at Month 30 (mg/dL)
Acoramidis
Acoramidis + Tafamidis
Placebo
Placebo + Tafamidis

Figure 7. *Phase 3 ATTRibute-CM results by concomitant tafamidis groups for mITT Population.*
A) *Mean change from baseline in serum TTR at Month 30.*
B) *Median of WB %Stabilization at Month 30.*
C) *Median of FPE %Stabilization at Month 30.*
D) *Waterfall plot of variant patients WB %Stabilization Month 30.*
E) *Waterfall plot of variant patients change from baseline in serum TTR at Month 30. Note: variant = mutant TTR genotype.*

CONCLUSIONS

- At its target therapeutic steady-state trough concentration, acoramidis achieved near-complete TTR stabilization across 18 unique TTR genotypes.
 - In the subset of paired samples, the stabilization was significantly greater for acoramidis than for tafamidis even at its peak clinical concentration.
 - This observation held across a range of destabilizing mutations, including a ~2-fold greater stabilization than tafamidis for the prevalent cardiomyopathic V122I variant.
- Results from the positive randomized, controlled Phase 3 ATTRibute-CM study are consistent with in vitro findings.
 - Acoramidis achieved near-complete stabilization in WT and ATTRv patients at Month 30 as measured by ex vivo WB and FPE assays.
 - Acoramidis achieved a greater degree of TTR stabilization as compared to clinically relevant concentrations of tafamidis, independent of TTR genotype.
- In vitro and in vivo assessments of TTR stabilization demonstrated acoramidis activity across 18 unique TTR variants encountered.

REFERENCES

1. Rapezzi, C., Quarta, C.C., Obici, L., et al. (2013). Disease profile and differential diagnosis of hereditary transthyretin-related amyloidosis with exclusively cardiac phenotype: an Italian perspective. Eur. Heart J. 34, 520–528.
2. Sekijima, Y., Wiseman, R.L., Matteson, … and Kelly, J.W. (2005). The biological and chemical basis for tissue-selective amyloid disease. Cell 121, 73–85.
3. Judge, D.P., Heitner, S.B., Falk, R.H., et al. (2019). Transthyretin Stabilization by AG10 in Symptomatic Transthyretin Amyloid Cardiomyopathy. J. Am. Coll. Cardiol. 74, 285–295.
4. Summary Review for Regulatory Action – NDA 211996/NDA212161 (tafamidis meglumine/free acid). May 2, 2019: p12.

bridgebio